Optimization of Polyethyleneimine Functionalized Spongy Monoliths for Lipid Nanoparticle Separations Using Liquid Chromatography – Mass Spectrometry

Brady Drennan[1], Phat Dinh[1], Rebecca Taylor[1], Kevin A. Schug[1], Samuel H. Yang[2]
[1]Department of Chemistry and Biochemistry, The University of Texas at Arlington, TX, USA
[2]Synthetic Molecule Pharmaceutical Sciences, Genentech, San Francisco, CA, USA

THE UNIVERSITY OF TEXAS AT ARLINGTON

Genentech
A Member of the Roche Group

Abstract

Lipid nanoparticles (LNPs) encapsulate therapeutic nucleotides to protect their labile structure and facilitate cellular uptake. Conventionally, dynamic light scattering (DLS) is used to determine average LNP size and size distribution. The main drawback of DLS is that it is a batch analytical technique, limiting its effectiveness for highly polydisperse samples or those with multiple size populations. For these more heterogeneous samples, separations are required prior to size determination. Size exclusion chromatography (SEC) is often used to combat this shortcoming. However, the analysis of LNPs on commercial SEC columns is not ideal due to insufficient pore sizes, shear forces, and over-adsorption of LNPs onto the stationary phases. In this study, spongy monoliths were modified with polyethyleneimine (PEI), and reaction conditions were optimized, including PEI molecular weight, pH, reaction time, and the addition of a cross-linking step. Optimized conditions demonstrated a reduction in LNP over-adsorption, with the amount of sample required to saturate the monolith reduced to 40 µL, compared to 400 µL used for an SEC column. Two distinct distributions of LNPs were also selectively separated, with the later-eluting distribution possessing four times more cholesterol than the earlier-eluting one.

Introduction

What are LNPs?

- Drug delivery vesicles
- Composed of ionizable lipids, cholesterol, PEGylated lipids, and phospholipids[1,2]

Drawbacks of previous separation methods that used Size Exclusion Chromatography (SEC)[3,4]:

- Conditioning and saturation of LNP on columns
- Secondary interaction between LNP and SEC columns

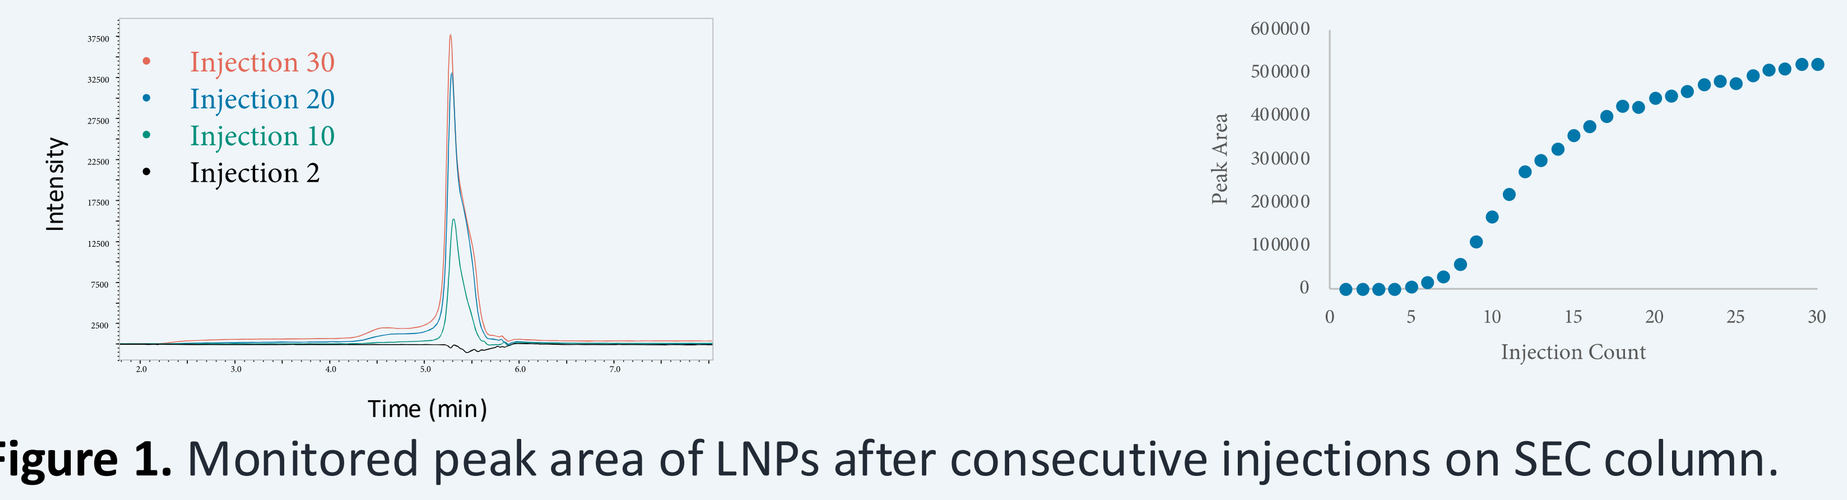
Injection 30
Injection 20
Injection 10
Injection 2
Intensity
Time (min)
Peak Area
Injection Count

Figure 1. Monitored peak area of LNPs after consecutive injections on SEC column.

Why spongy monolith columns?[5]

- Low back pressure due to large pore size
- Modifiable with different functional groups
- Low adsorption due to low surface area

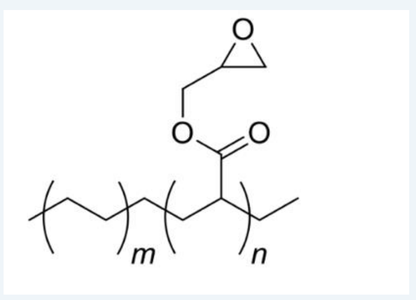

poly(ethylene-co-glycidyl methacrylate) (PEGM)
Amount of Epoxy Group: 12%

Why PEI?

- Decreases electrostatic interaction between LNP and column
- Used in capillary electrophoresis (CE) to separate proteins

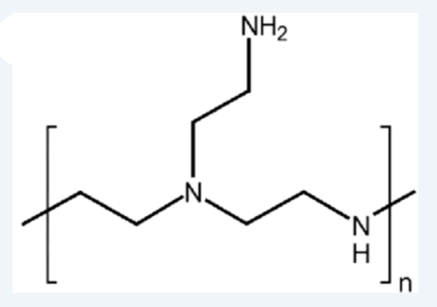

Polyethyleneimine (PEI), branched

Goal:

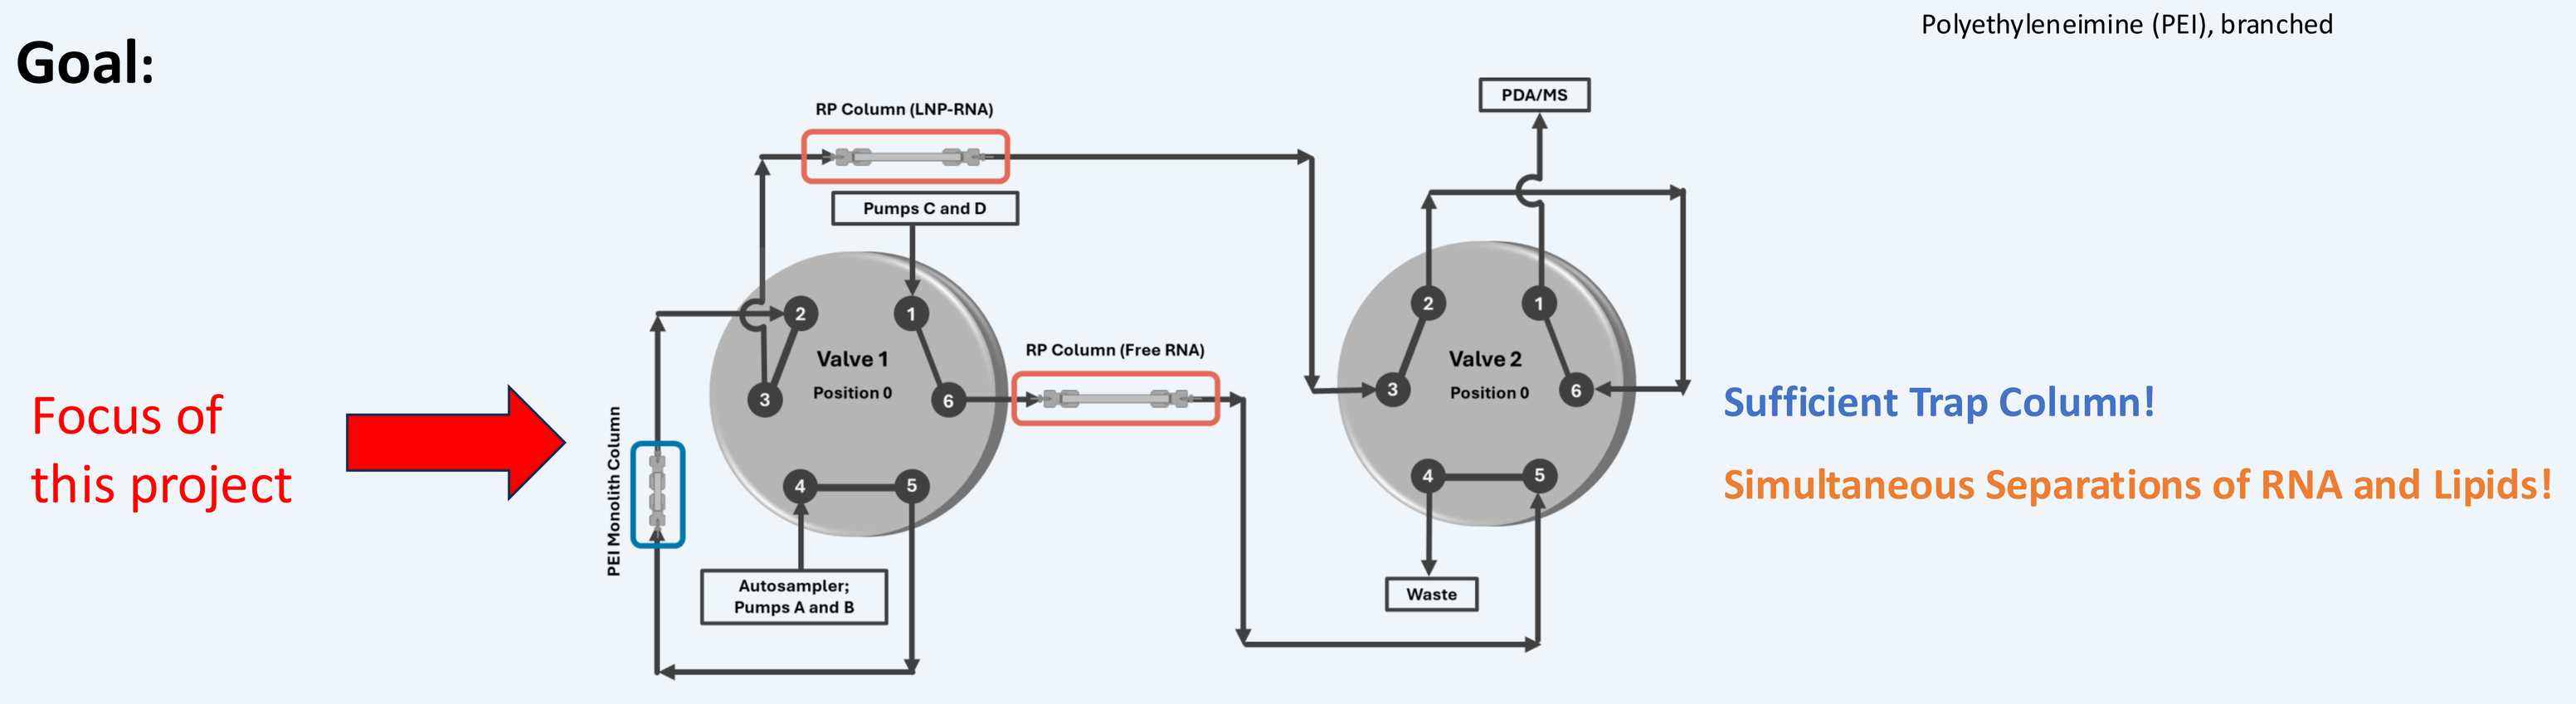
Focus of this project
RP Column (LNP-RNA)
Pumps C and D
Valve 1
Position 0
RP Column (Free RNA)
PDA/MS
Valve 2
Position 0
PEI Monolith Column
Autosampler; Pumps A and B
Waste
Sufficient Trap Column!
Simultaneous Separations of RNA and Lipids!

Figure 2: Online Multimodal LC-MS System for Encapsulation Efficiency Determination

Methodology

Empty LNP Synthesis Procedures[6]

1. Reagent Preparation
 - Ethanolic lipid mixture
 - Aqueous buffer: 50 mM sodium acetate, no RNA cargo

2. Rapid Mixing: Pipette Technique
 - 3:1 (aqueous : ethanolic)

3. Dialysis in PBS x3, MWCO: 10 kDa

NOTE: "Heavy"- LNP made by replacing DSPC with DSPC-d70.

Lipid Mixture Component	Stock Solutions		Working Mixture		
	mg/ml	MW	Molar Ratio	mg	Required Volume
SM-102	100	710.2	50	3.55	35 µl
1,2-DSPC	25	790.2	10	0.79	32 µl
Cholesterol	5	386.7	38.5	1.48	296 µl
DMG-PEG(2000)	1	2,526	1.5	0.38	378 µl
Absolute ethanol					259 µl
Total				6.20	1 ml

Figure 3: Ethanolic Lipid Mixture Preparation

PEI Functionalized Monoliths General Synthesis Procedures

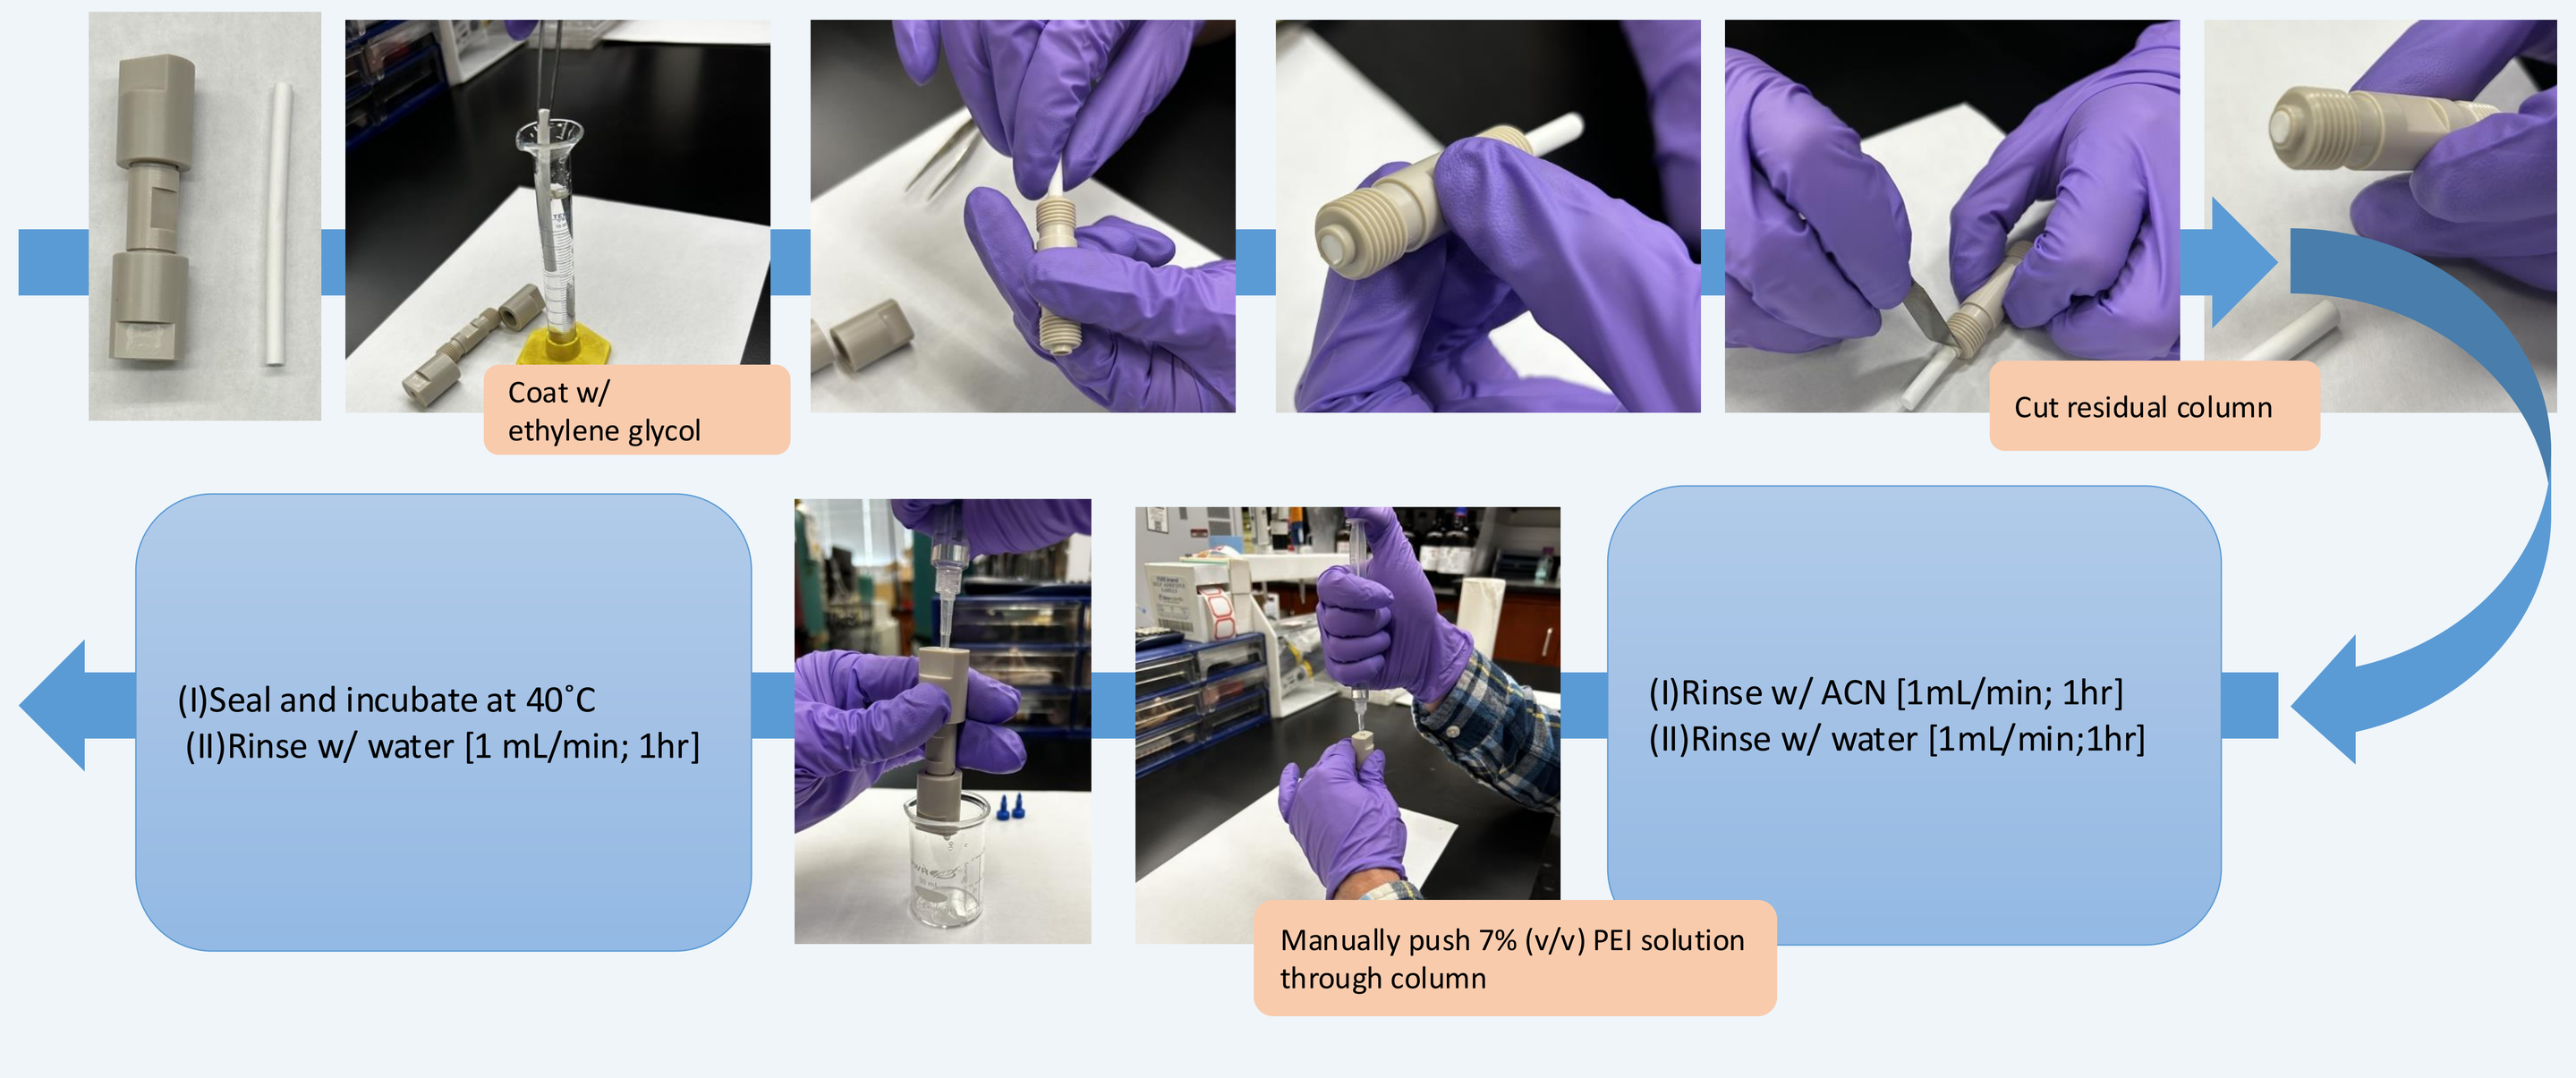
Coat w/ ethylene glycol
Cut residual column
(I)Rinse w/ ACN [1mL/min; 1hr]
(II)Rinse w/ water [1mL/min;1hr]
Manually push 7% (v/v) PEI solution through column
(I)Seal and incubate at 40˚C
(II)Rinse w/ water [1 mL/min; 1hr]

Results

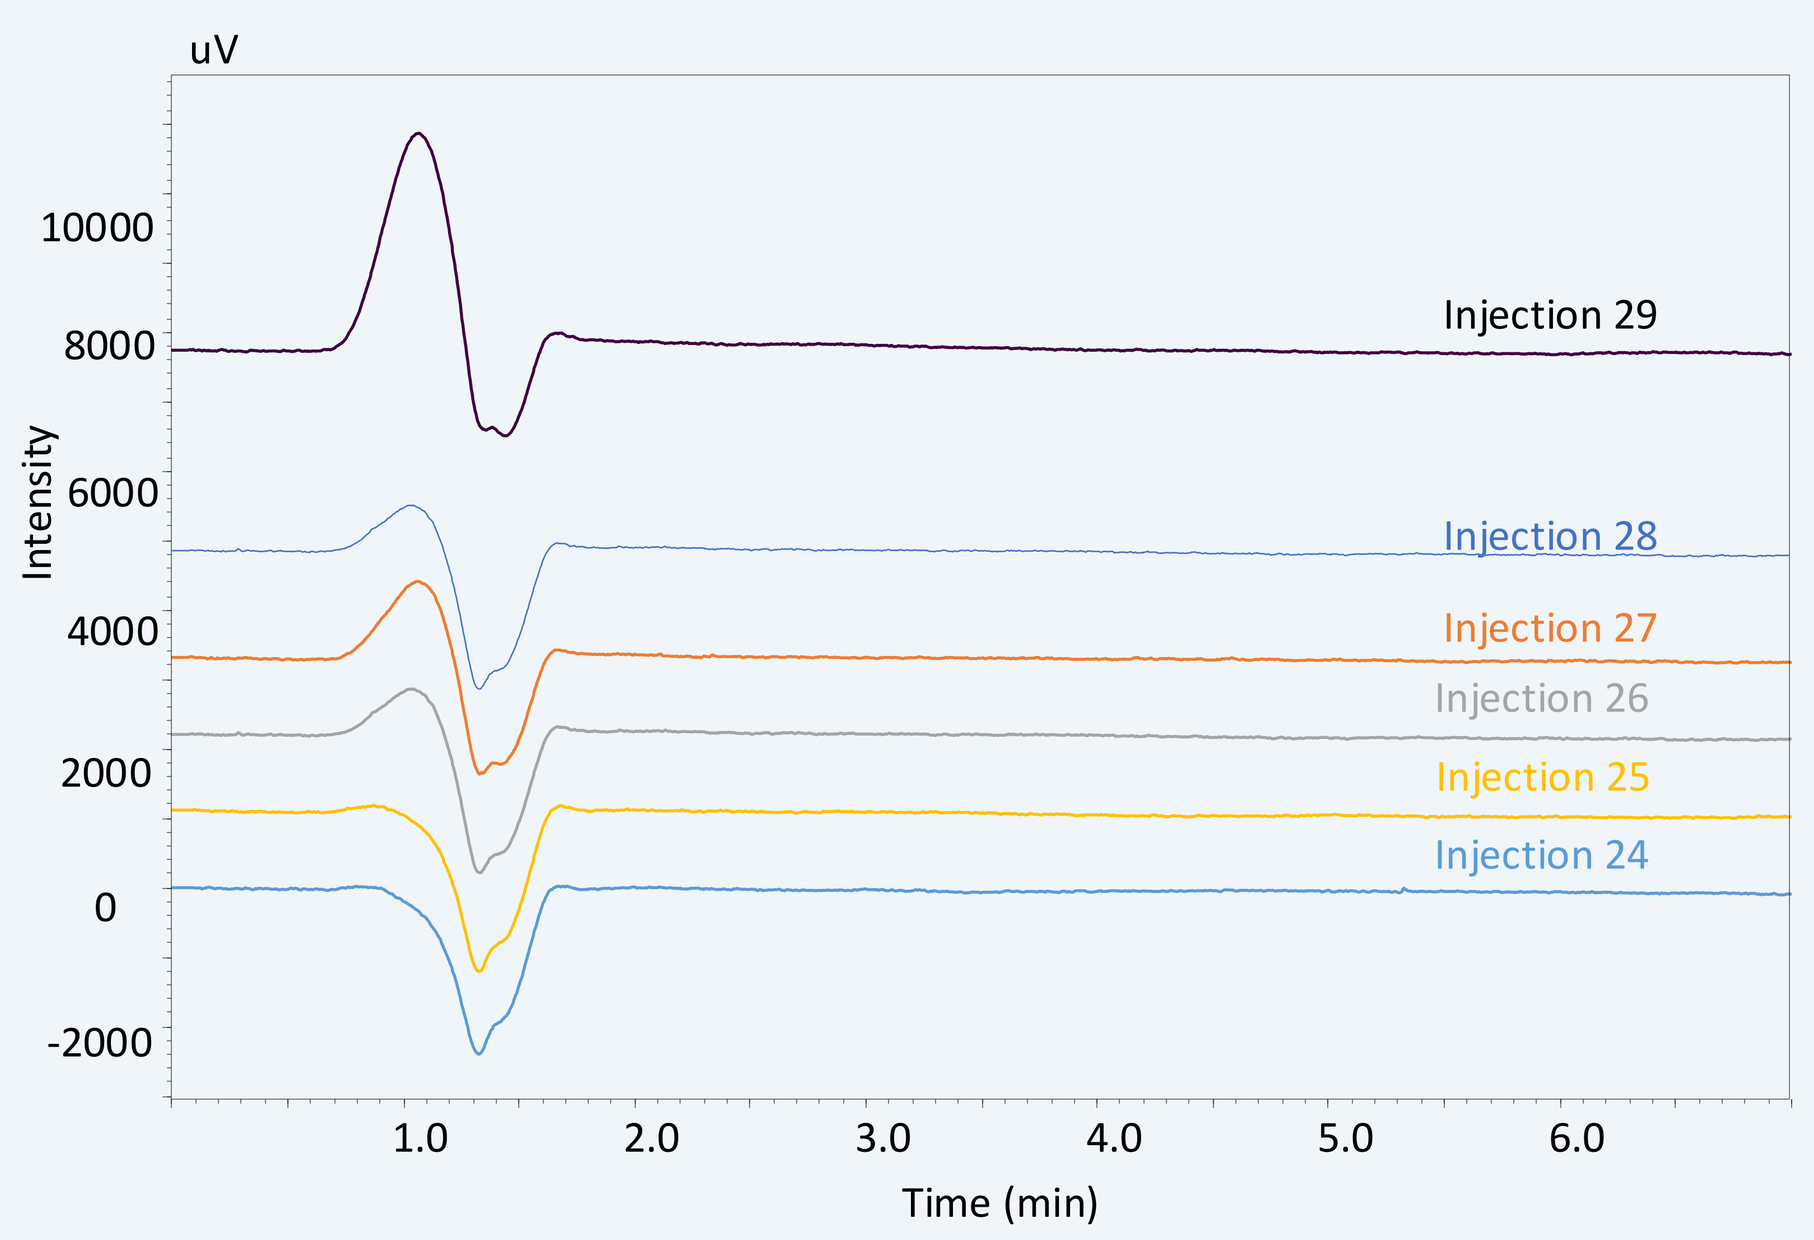
uV
Intensity
Injection 29
Injection 28
Injection 27
Injection 26
Injection 25
Injection 24
Time (min)

Figure 4: LNP adsorption on 600 Da functionalized monolith, 24 hr reaction time

Column:
600 MW PEI Modified Monolith
(4.6 mm i.d. x 50 mm, Pore Size: 30 µm)
Reaction Conditions:
7% PEI, 40˚C, 24hr
Mobile Phase:
A: 50 mM Ammonium Acetate
Injection Volume:
10 µL, neat
Time Program:
100% A, Isocratic (7 min)
Flow Rate:
0.50 mL/min
Detection:
PDA, 254 nm

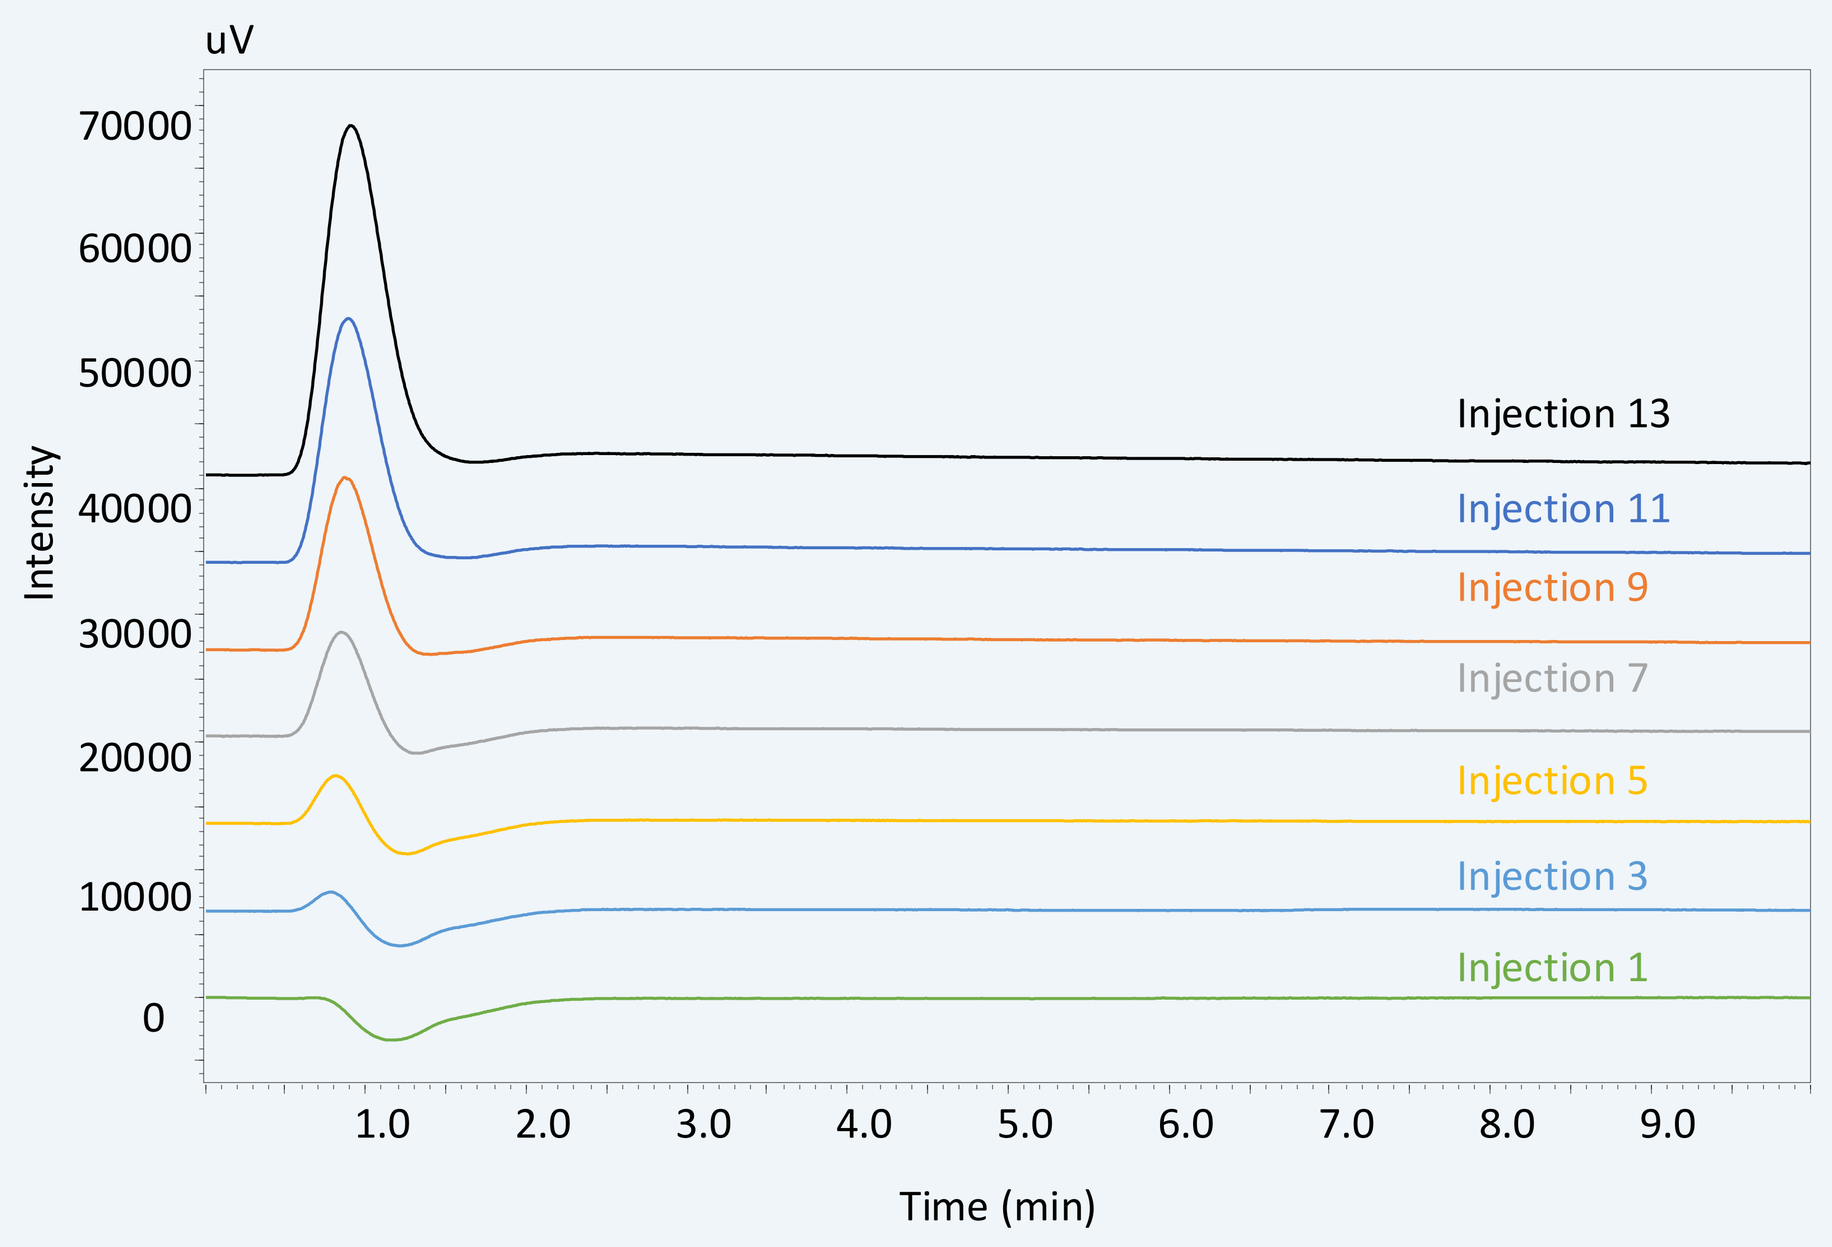
uV
Intensity
Injection 13
Injection 11
Injection 9
Injection 7
Injection 5
Injection 3
Injection 1
Time (min)

Figure 5: LNP adsorption on 2 kDa functionalized monolith, 24 hr reaction time

Column:
2 kDa PEI Modified Monolith
(4.6 mm i.d. x 50 mm, Pore Size: 30 µm)
Reaction Conditions:
7% PEI, 40˚C, 24hr
Mobile Phase:
A: 50 mM Ammonium Acetate
Injection Volume:
10 µL, neat
Time Program:
100% A, Isocratic (10 min)
Flow Rate:
0.50 mL/min
Detection:
PDA, 254 nm

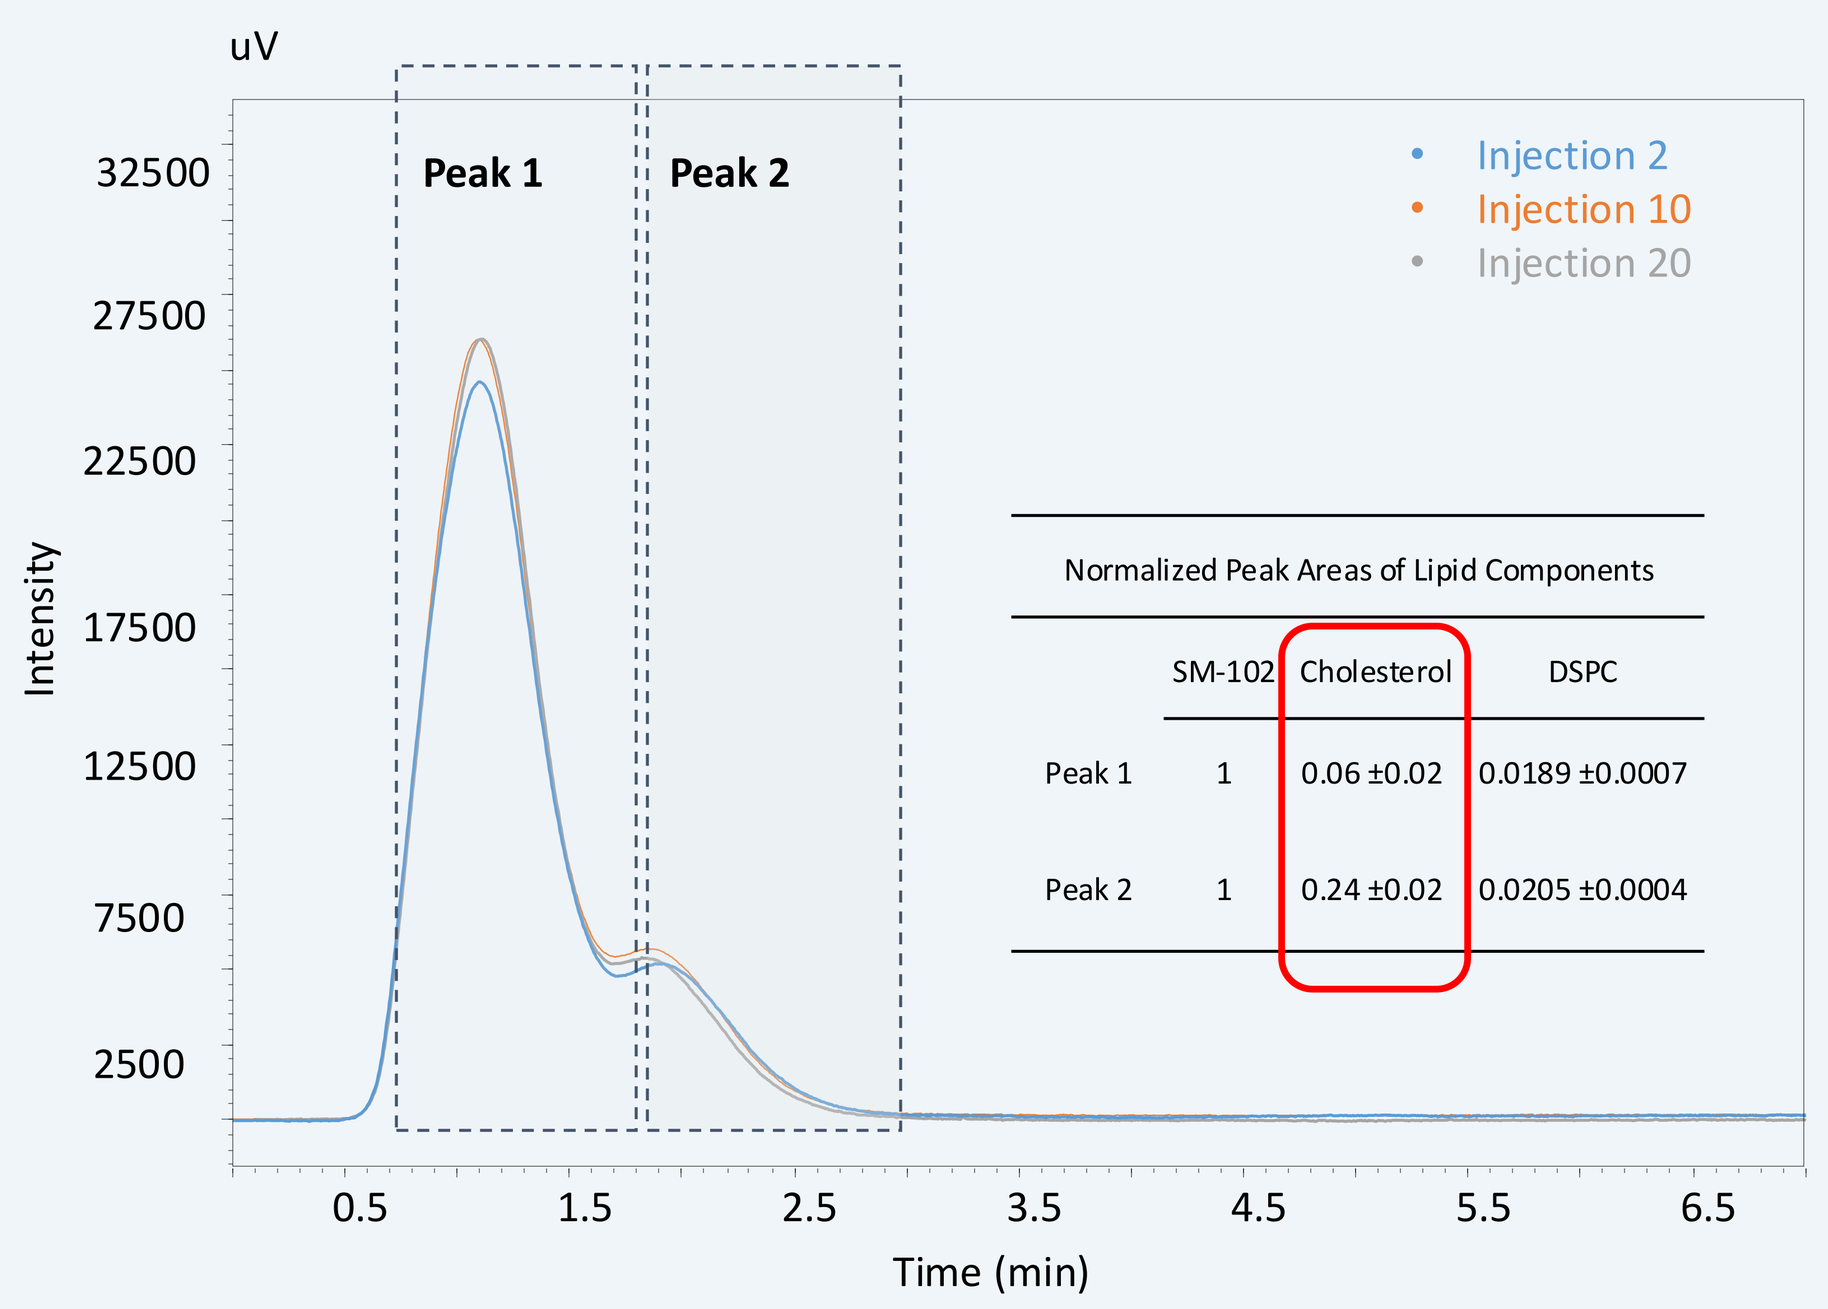
uV
Intensity
Peak 1
Peak 2
Injection 2
Injection 10
Injection 20
Normalized Peak Areas of Lipid Components
SM-102 Cholesterol DSPC
Peak 1 1 0.06 ±0.02 0.0189 ±0.0007
Peak 2 1 0.24 ±0.02 0.0205 ±0.0004
Time (min)

Figure 6: LNP adsorption on 2 kDa functionalized monolith, 24 hr reaction time, crosslinked with EDGE on PDA

Column:
2 kDa MW, Cross-linked PEI Modified Monolith
(4.6 mm i.d. x 50 mm, Pore Size:30 µm)
Reaction Conditions:
7% PEI, 0.1% TEA (pH 11), 10 mL
40˚C, 24hr → 70% EDGE, 0.1% TEA (pH 11), MeOH, 5mL
Mobile Phase:
A: 0.1% FA, pH ~3
Injection Volume:
10 µL, neat
Time Program:
100% A, Isocratic (7 min)
Flow Rate:
0.50 mL/min
Detection:
PDA, 254 nm

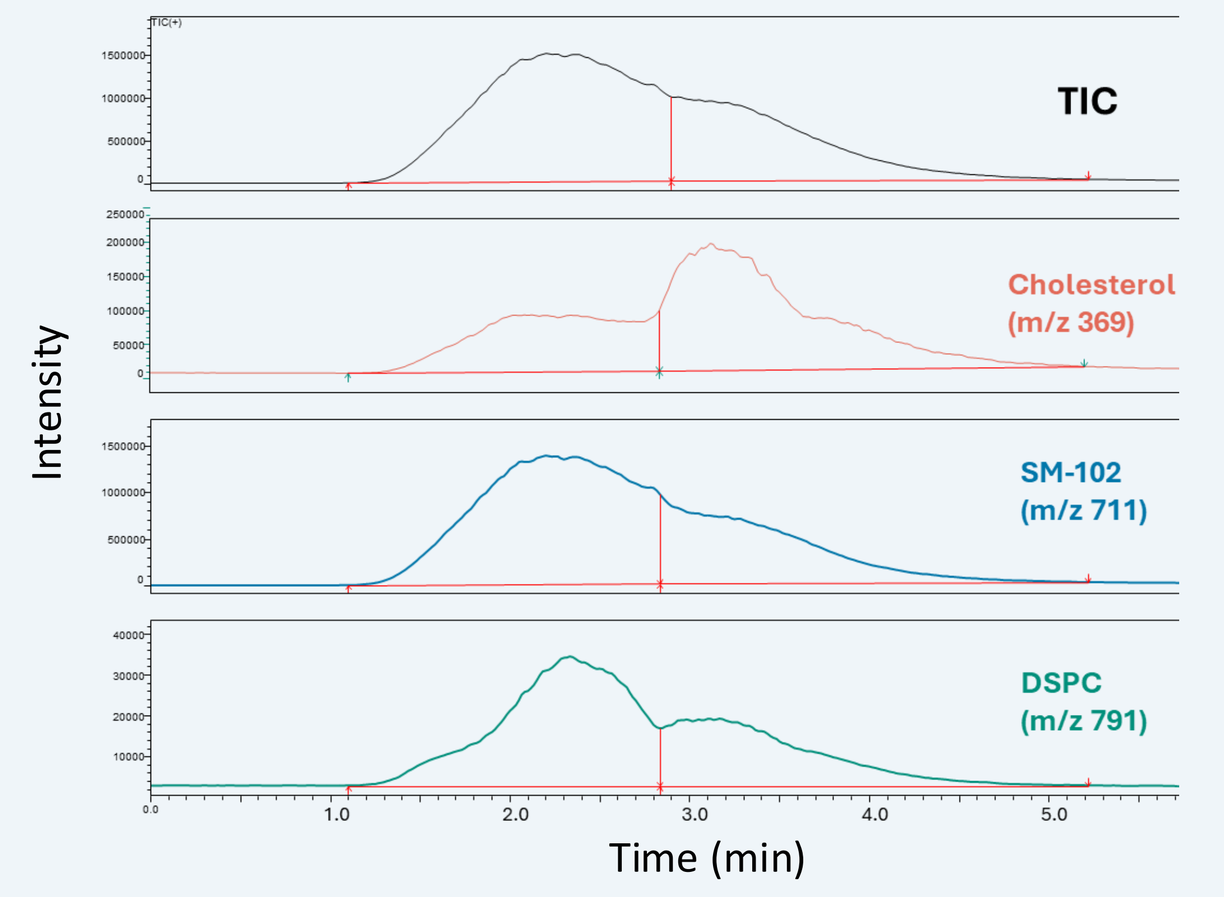
TIC
Cholesterol (m/z 369)
SM-102 (m/z 711)
DSPC (m/z 791)
Intensity
Time (min)

Figure 7: LNP adsorption on 2 kDa functionalized monolith, 24 hr reaction time, crosslinked with EDGE on MS

**decreasing flow rate for ESI (0.25 mL/min), reducing resolution*

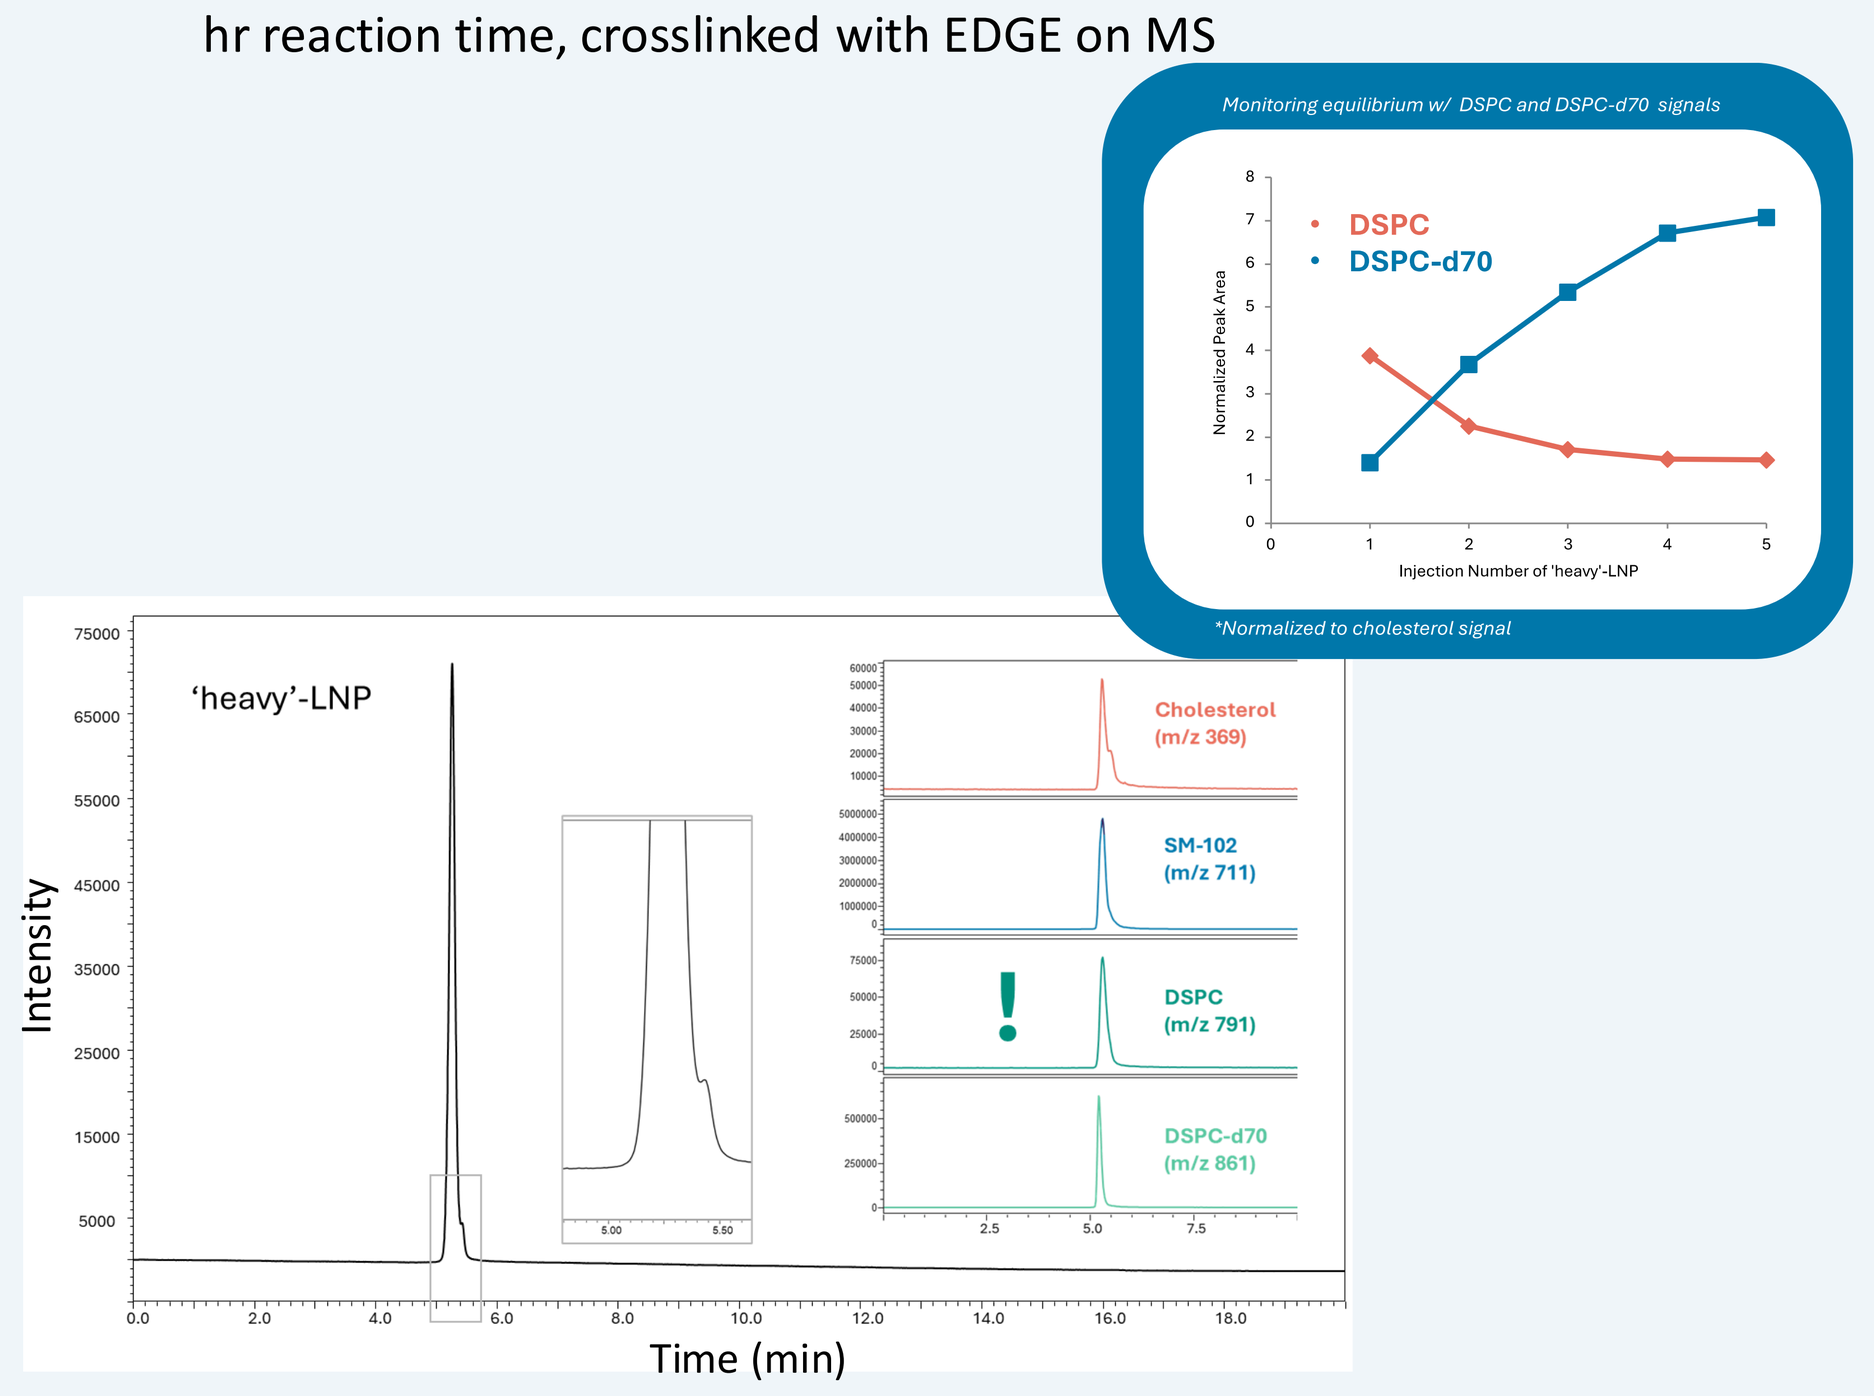
Monitoring equilibrium w/ DSPC and DSPC-d70 signals
DSPC
DSPC-d70
Normalized Peak Area
Injection Number of 'heavy'-LNP
*Normalized to cholesterol signal
'heavy'-LNP
Intensity
Cholesterol (m/z 369)
SM-102 (m/z 711)
DSPC (m/z 791)
DSPC-d70 (m/z 861)
Time (min)

Figure 8: DPSC-d70 in equilibrium with DSPC

Column:
MacMod, 'Monodisperse' SEC-1000
(3µm, 4.6 x 150 mm)
Mobile Phase:
A: 10 mM Ammonium Formate, 0.1% FA
Injection Volume:
10 µL, neat
Time Program:
100% A, Isocratic (20 min)
Flow Rate:
0.40 mL/min
Detection:
PDA, 254 nm
1Q-MS, SIM (+); m/z 369, 711, 791

Conclusions

- PEI molecular weight, pH, reaction time, and addition of a cross-linking step all contributed to the reduction of LNP adsorption onto the stationary phase.
- Two distinct compositional distributions of LNP were observed and separated on the monolith, with the later-eluting distribution possessing four times more cholesterol than the earlier-eluting one.
- Separations of LNP compositional distributions have only been done on CE, and this fast separation is done on LC, ideal for high throughput experiments.
- Coupling the LC with MS is also very common and easy, ideal for optimization of LNP formulations and quality control purposes.

Future Works

- Optimization of endcapping step to increase surface coverage.
- Column characterizations using elemental analysis, frontal analysis, and scanning electron microscope.
- Profile retention with an analyte test mix that is composed of RNAs, proteins, and small molecules.

References

1. Metkar et al., Nat. Rev. Drug Discov. **2024**, 23, 67.
2. Hou et al., *Nat. Rev. Mater.* **2021**, 6, 1078-1094.
3. Zhang et al., *Anal. Chem.* **2012**, 84, 6088–6096.
4. Grabielle-Madelmont et al., *J Biochem Biophys Methods.* **2003**, 56, 189-217.
5. Kubota et al., *Nat. Sci. Rep.* **2017**, 7, 178.
6. *LipidLaunch LNP-102 Exploration Kit, Caymen Chemical, 2024.*

Acknowledgments

The authors acknowledge the support and guidance from the University of Texas at Arlington, Shimadzu Inc., Spongy Monoliths (SPM) and MAC-MOD Analytical, Inc.

SHIMADZU Excellence in Science
SPM Separation & Purification Maneuver
macmod SMARTER CHROMATOGRAPHY